MON-594

ENDO2025
JULY 12–15, 2025 SAN FRANCISCO, CA

JUV-161 Improves Glucose Metabolism and Muscle Health in Type 2 Diabetes Mice

Vengadeshprabhu Karuppagounder, Ritwik Datta, HeeJu Kim, Ashil Koranne, Annie Yang, Sharon Louie, Han Song, Zhihua Li, Thach Mai, Banmeet Anand, Priya Handa, Jeremy D. O'Connell, Hanadie Yousef

**The authors have no conflicts. They are employees of Juvena Therapeutics.*

Introduction

- **Type 2 diabetes** remains a major global contributor to morbidity, mortality, and healthcare burden.
- GIPR/GLP-1R agonists have led to significant clinical advances, improving glycemic control and reducing body weight, largely through appetite suppression and enhanced insulin secretion.
- However, these benefits are often accompanied by unintended lean mass loss, raising concerns about long-term metabolic health, frailty risk, and treatment sustainability.
- There remains a high unmet medical need for:
 - Durable glycemic control across heterogeneous populations
 - Therapies with novel or complementary mechanisms of action
 - Strategies that preserve lean body mass while improving metabolic outcomes
 - Alternatives for patients with suboptimal response or tolerability issues
- Next-generation therapies targeting distinct metabolic pathways are essential to broaden treatment options and optimize long-term disease management in type 2 diabetes.

Objective

To evaluate the therapeutic efficacy of JUV-161, a novel investigational compound, in improving glycemic control in the db/db mouse model of type 2 diabetes. Specifically, the study aims to assess the effects of JUV-161 on fasting blood glucose levels and glycated hemoglobin (HbA1c) over a defined treatment period. Secondary objectives include evaluation of insulin sensitivity, lean mass preservation, and the compound's potential to concurrently improve systemic metabolic parameters in obese type 2 diabetic mice.

Methods

This study was conducted using male db/db mice, a well-established model for obesity and type 2 diabetes. The treatment period lasted 4 weeks, during which mice were assigned to receive either vehicle control or JUV-161. To evaluate insulin sensitivity, an insulin tolerance test (ITT) was performed during week 3 of treatment. Mice received a single injection of JUV-161, followed by a 6-hour fast, and then an intraperitoneal (IP) injection of insulin. Blood glucose levels were measured over a 120-minute period following insulin administration, with the 0-minute time point representing the pre-insulin baseline. Additional metabolic assessments included fasting blood glucose and hemoglobin A1c (HbA1c), which were measured at baseline and at the end of the study following chronic JUV-161 treatment to assess improvements in glycemic control.

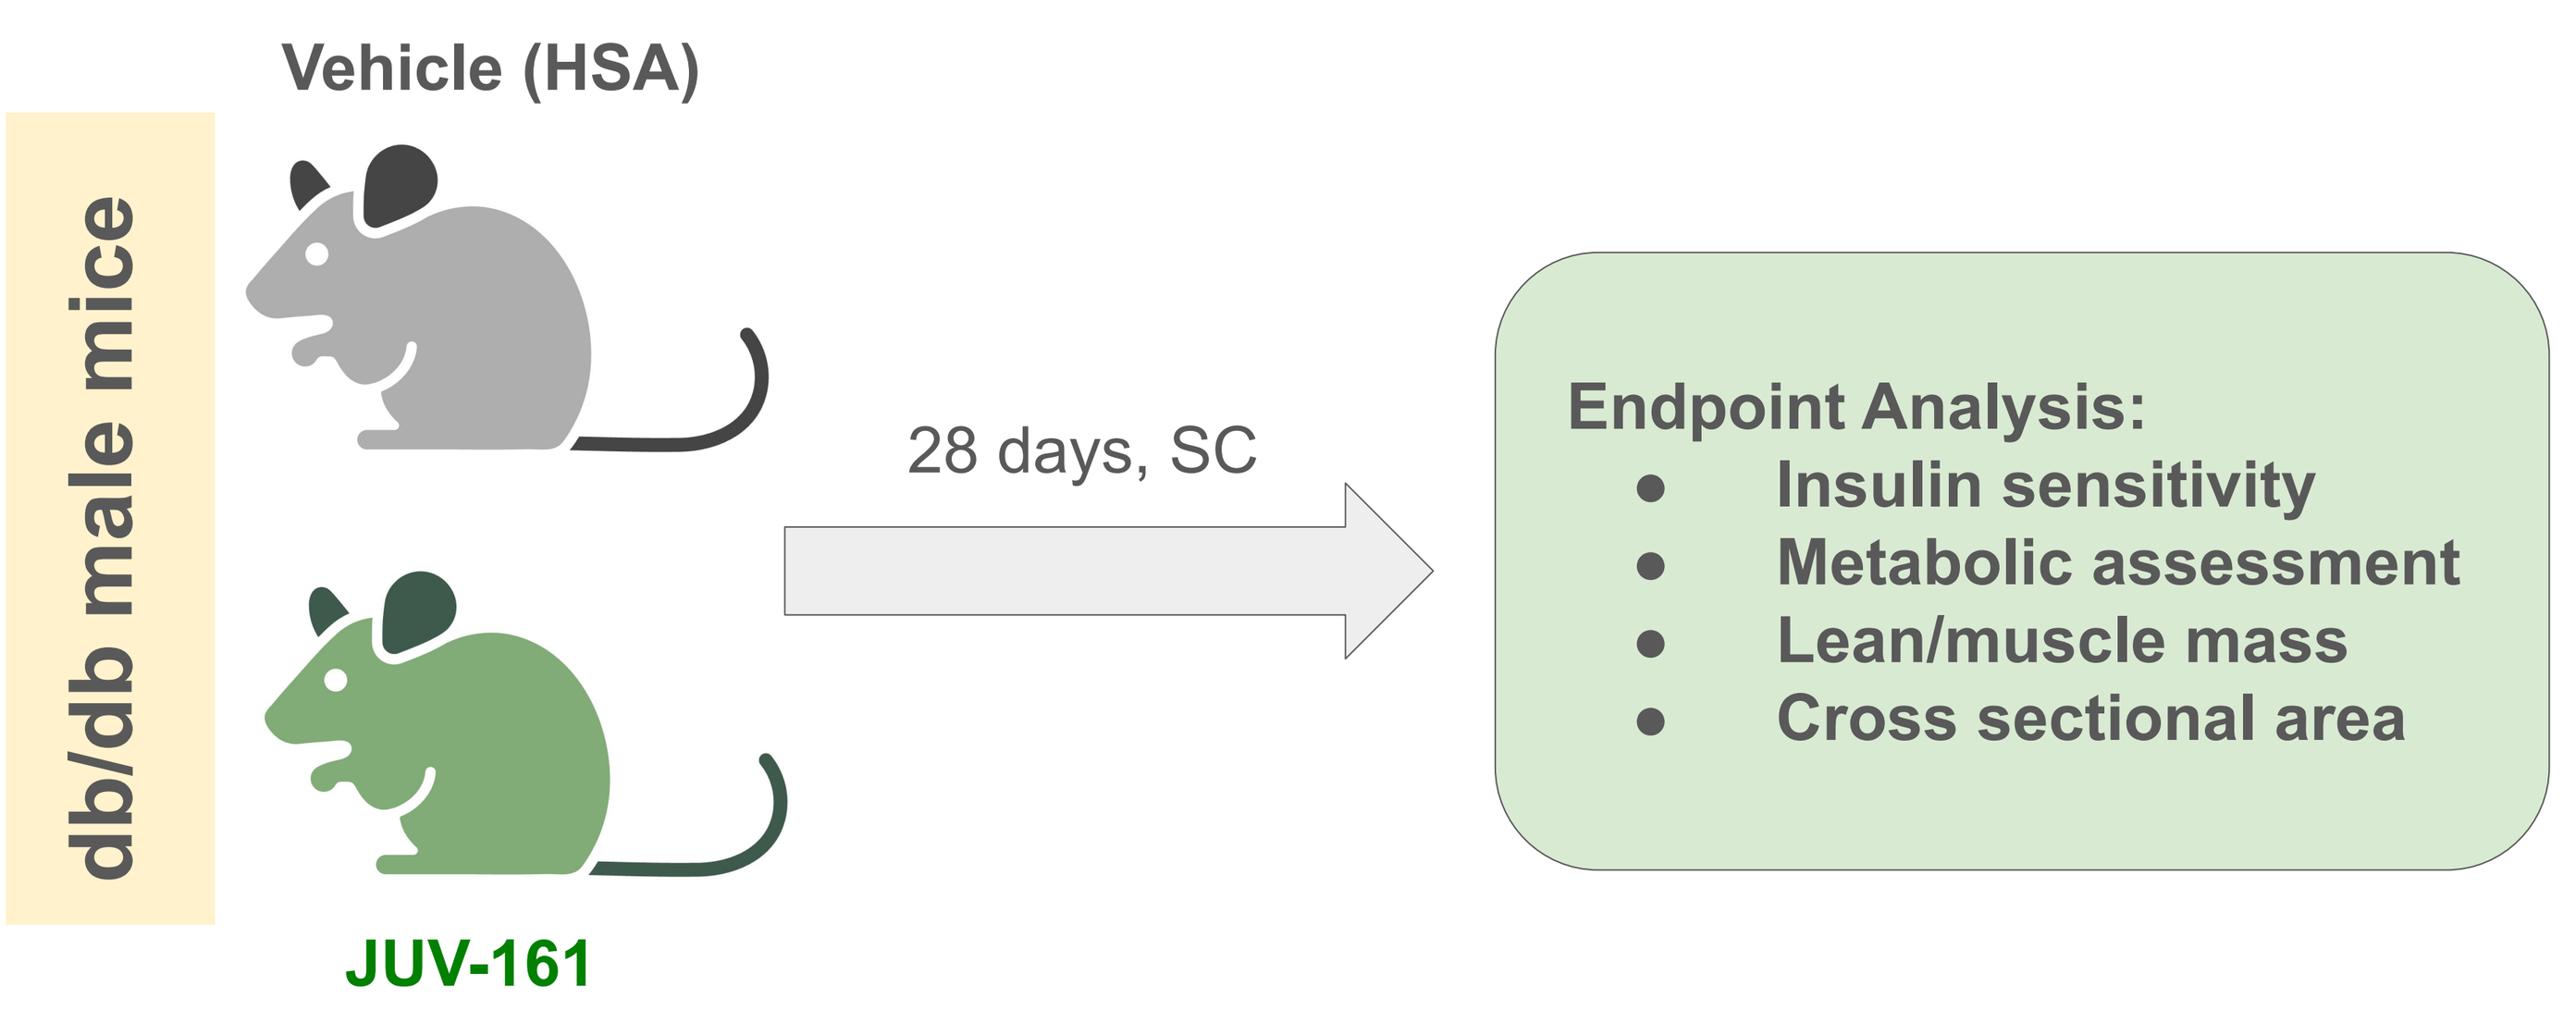
Vehicle (HSA)
db/db male mice
JUV-161
28 days, SC
Endpoint Analysis:
Insulin sensitivity
Metabolic assessment
Lean/muscle mass
Cross sectional area

Data Analysis: Data are presented as mean ± SEM. Statistical analyses were performed using two-way ANOVA followed by Dunnett's or Bonferroni's multiple comparison test for comparisons involving more than two groups. An unpaired t-test with Welch's correction was used for comparisons between two groups. Statistical significance is indicated as follows: ****p < 0.0001; ***p < 0.001; **p < 0.01; *p < 0.05.

Results

JUV-161 improves insulin sensitivity in obese type 2 diabetic mice

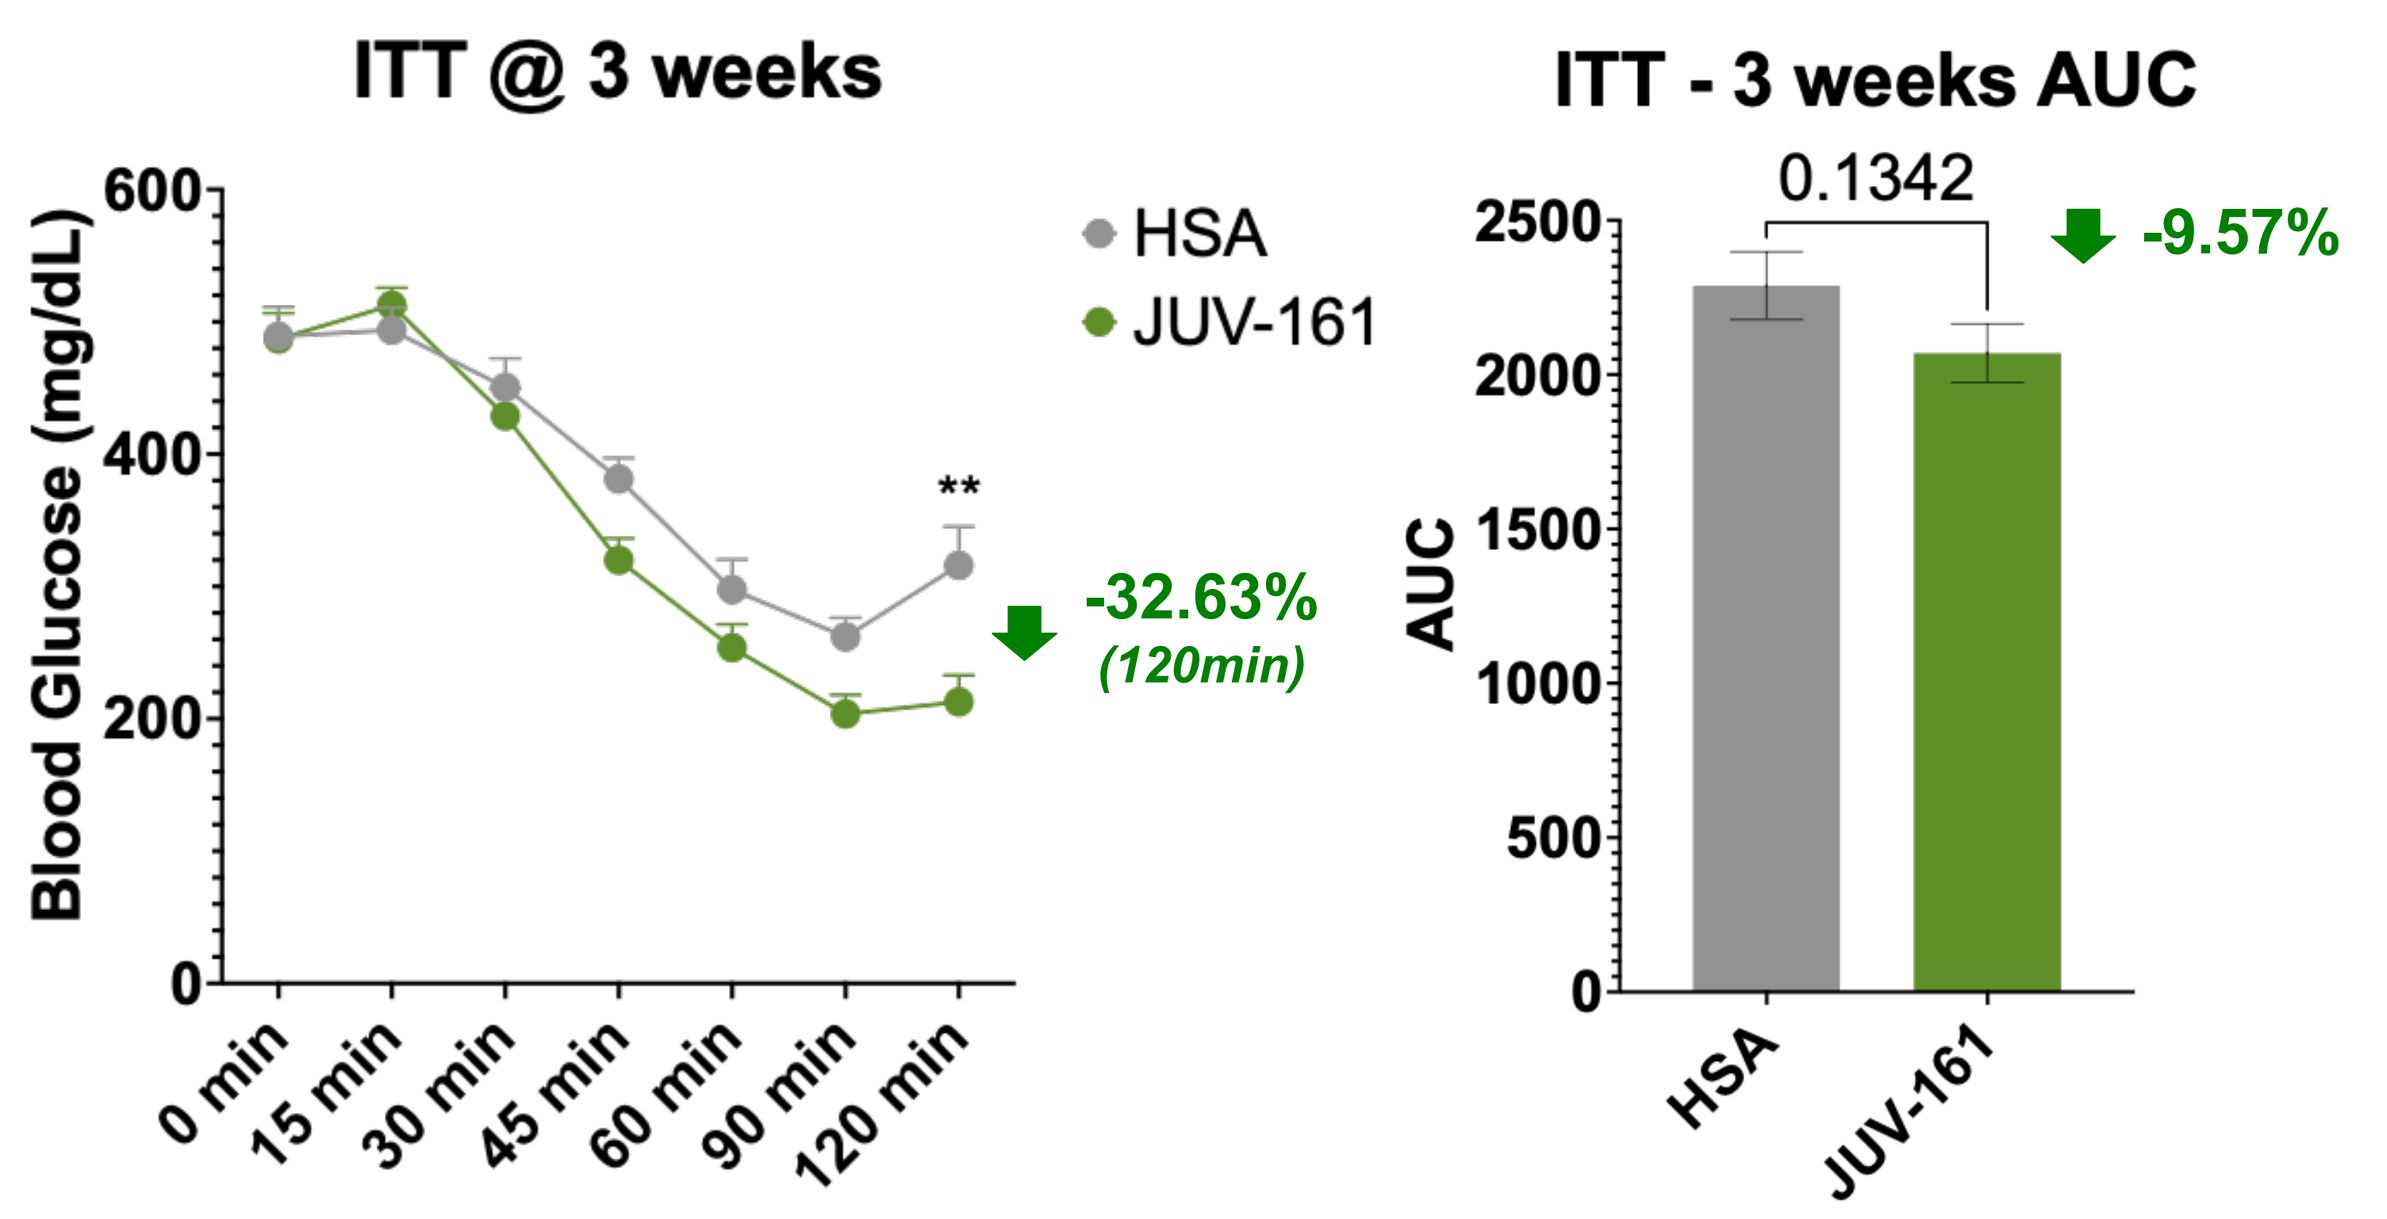
ITT @ 3 weeks
Blood Glucose (mg/dL)
HSA
JUV-161
-32.63%
(120min)
0 min
15 min
30 min
45 min
60 min
90 min
120 min
ITT - 3 weeks AUC
0.1342
-9.57%
AUC
HSA
JUV-161

Figure 1: Chronic JUV-161 treatment improves insulin sensitivity in db/db mice. *At the end of 3 weeks of chronic JUV-161 treatment, mice were dosed with JUV-161 and fasted for 6 hours. Mice were then injected with insulin and subjected to RO blood collection. Blood glucose levels were measured for the next 120 minutes. 0 minute represents baseline, prior to insulin injection, post 6 hrs fasting and injection of JUV-161. Two-way ANOVA followed by Dunnett's multiple comparison test. Statistical significance is indicated as follows**p<0.01*

JUV-161 improves glucose regulation in obese type 2 diabetic mice

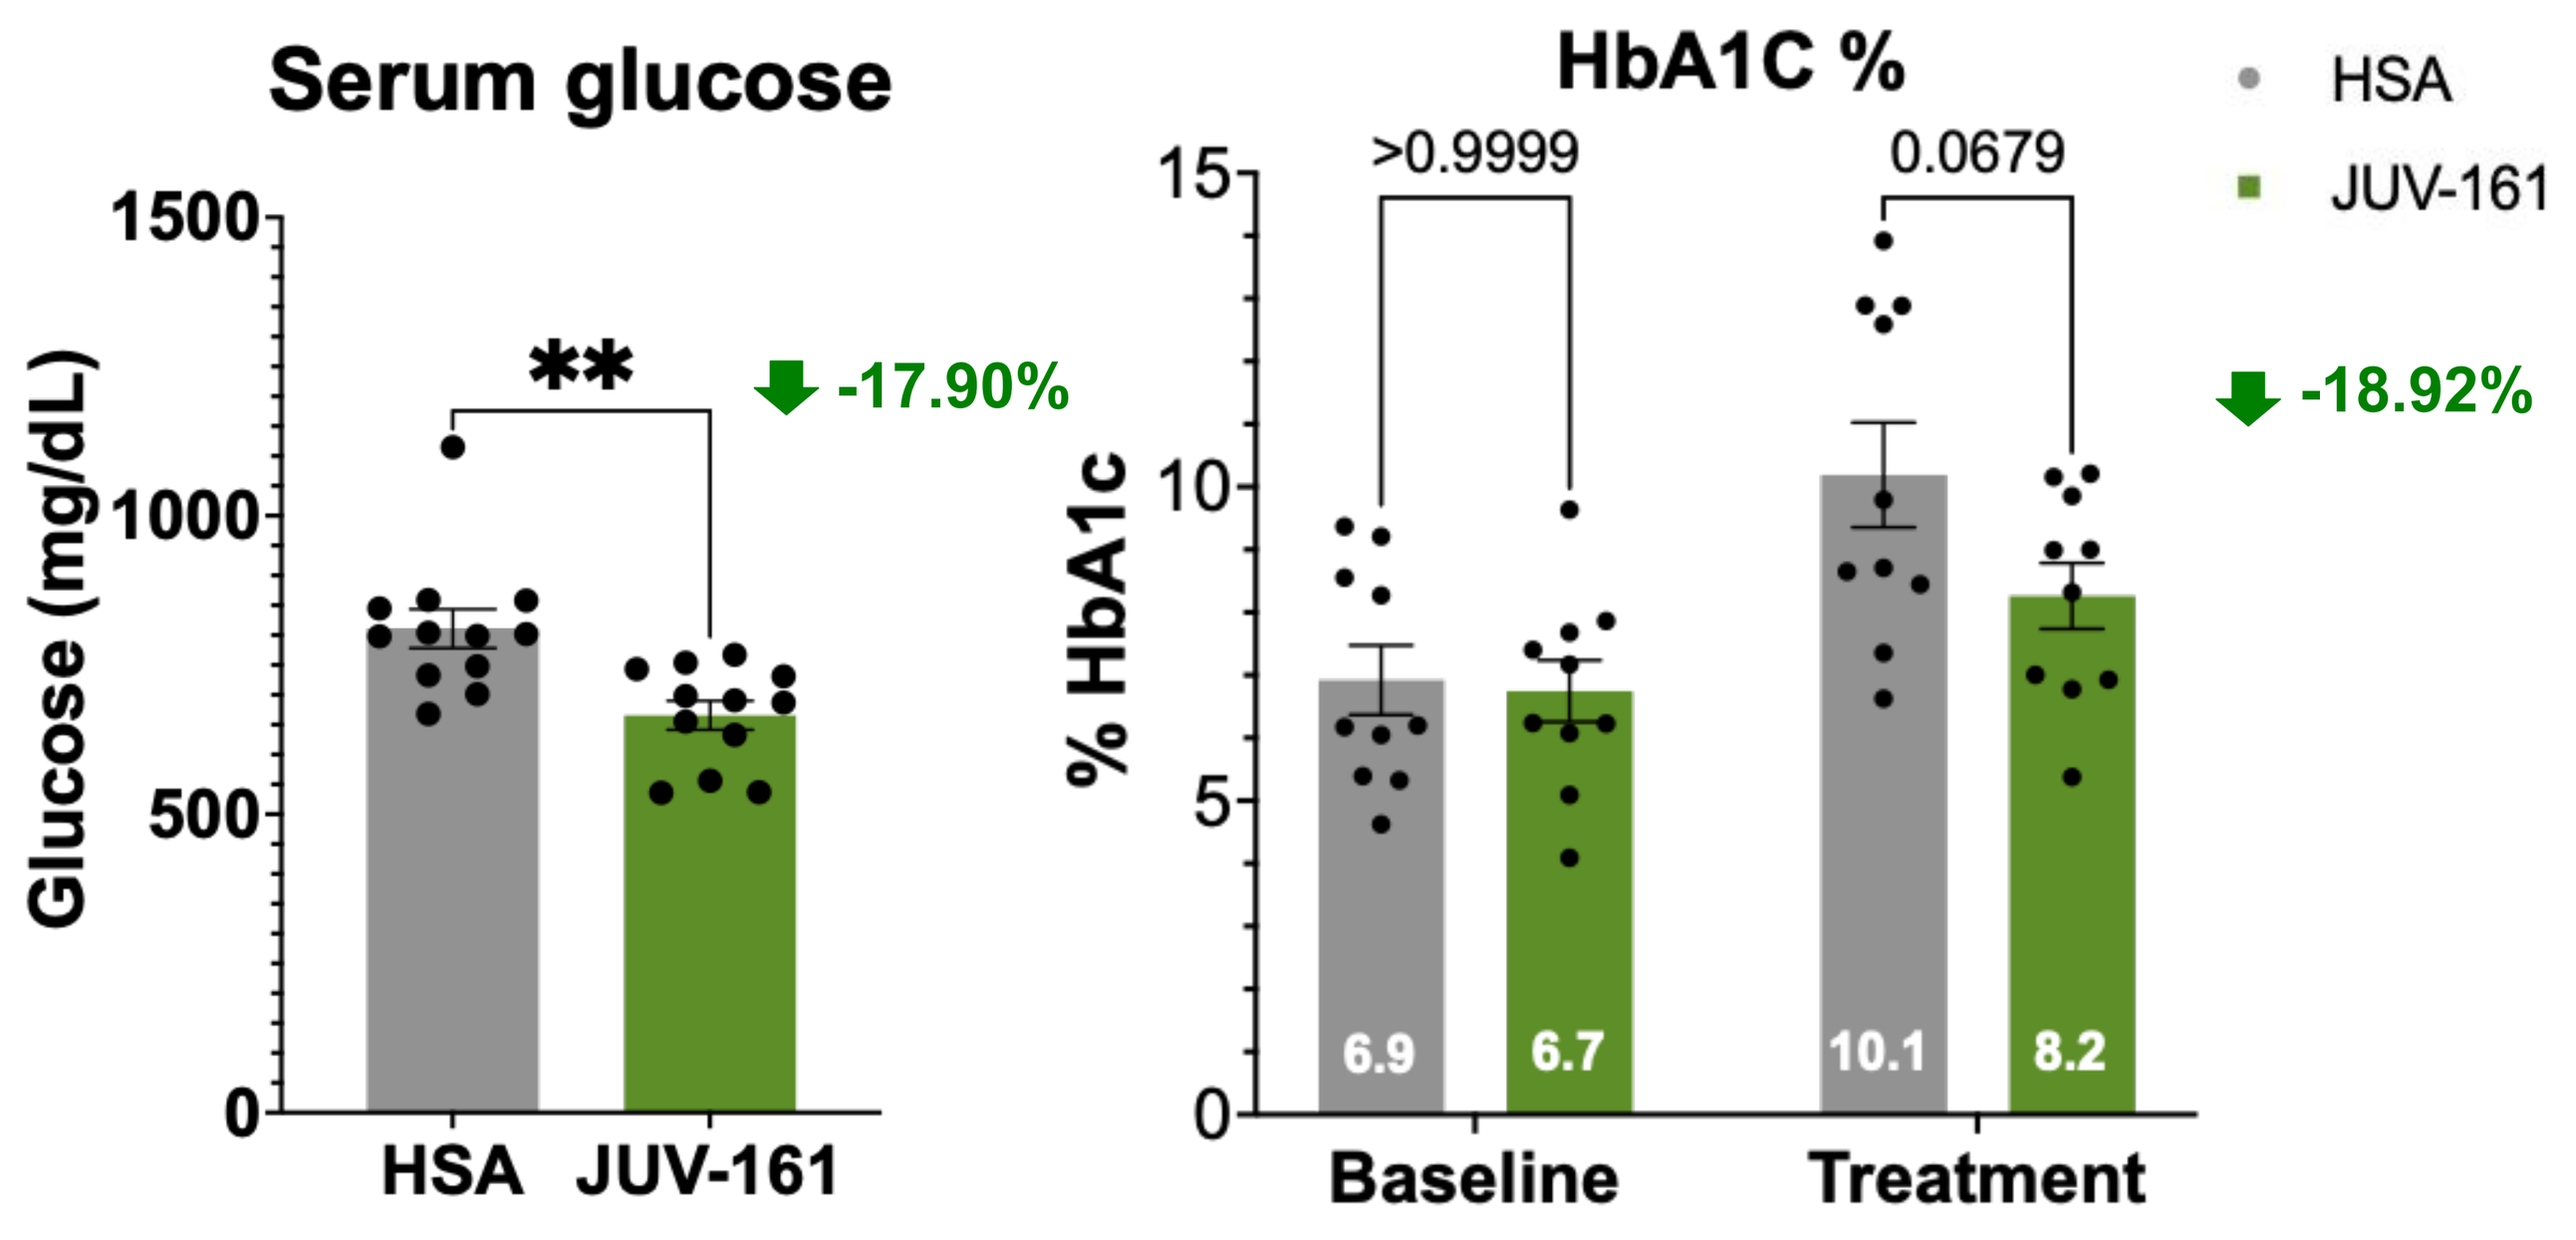
Serum glucose
Glucose (mg/dL)
-17.90%
HSA
JUV-161
HbA1C %
% HbA1c
>0.9999
0.0679
-18.92%
6.9
6.7
10.1
8.2
Baseline
Treatment

Figure 2: Chronic JUV-161 treatment improves glucose regulation in obese type 2 diabetic db/db mice. *At the end of the study, whole blood was collected for HbA1c (%) analysis, and serum was used to assess additional metabolic parameters. Statistical analysis was performed using unpaired t-tests with Welch's correction or two-way ANOVA followed by Bonferroni's multiple comparison test, as appropriate. Statistical significance is indicated as follows: **p < 0.01. Sample size: n = 12 per group.*

JUV-161 improves concurrently improve systemic metabolic parameters in obese type 2 diabetic mice

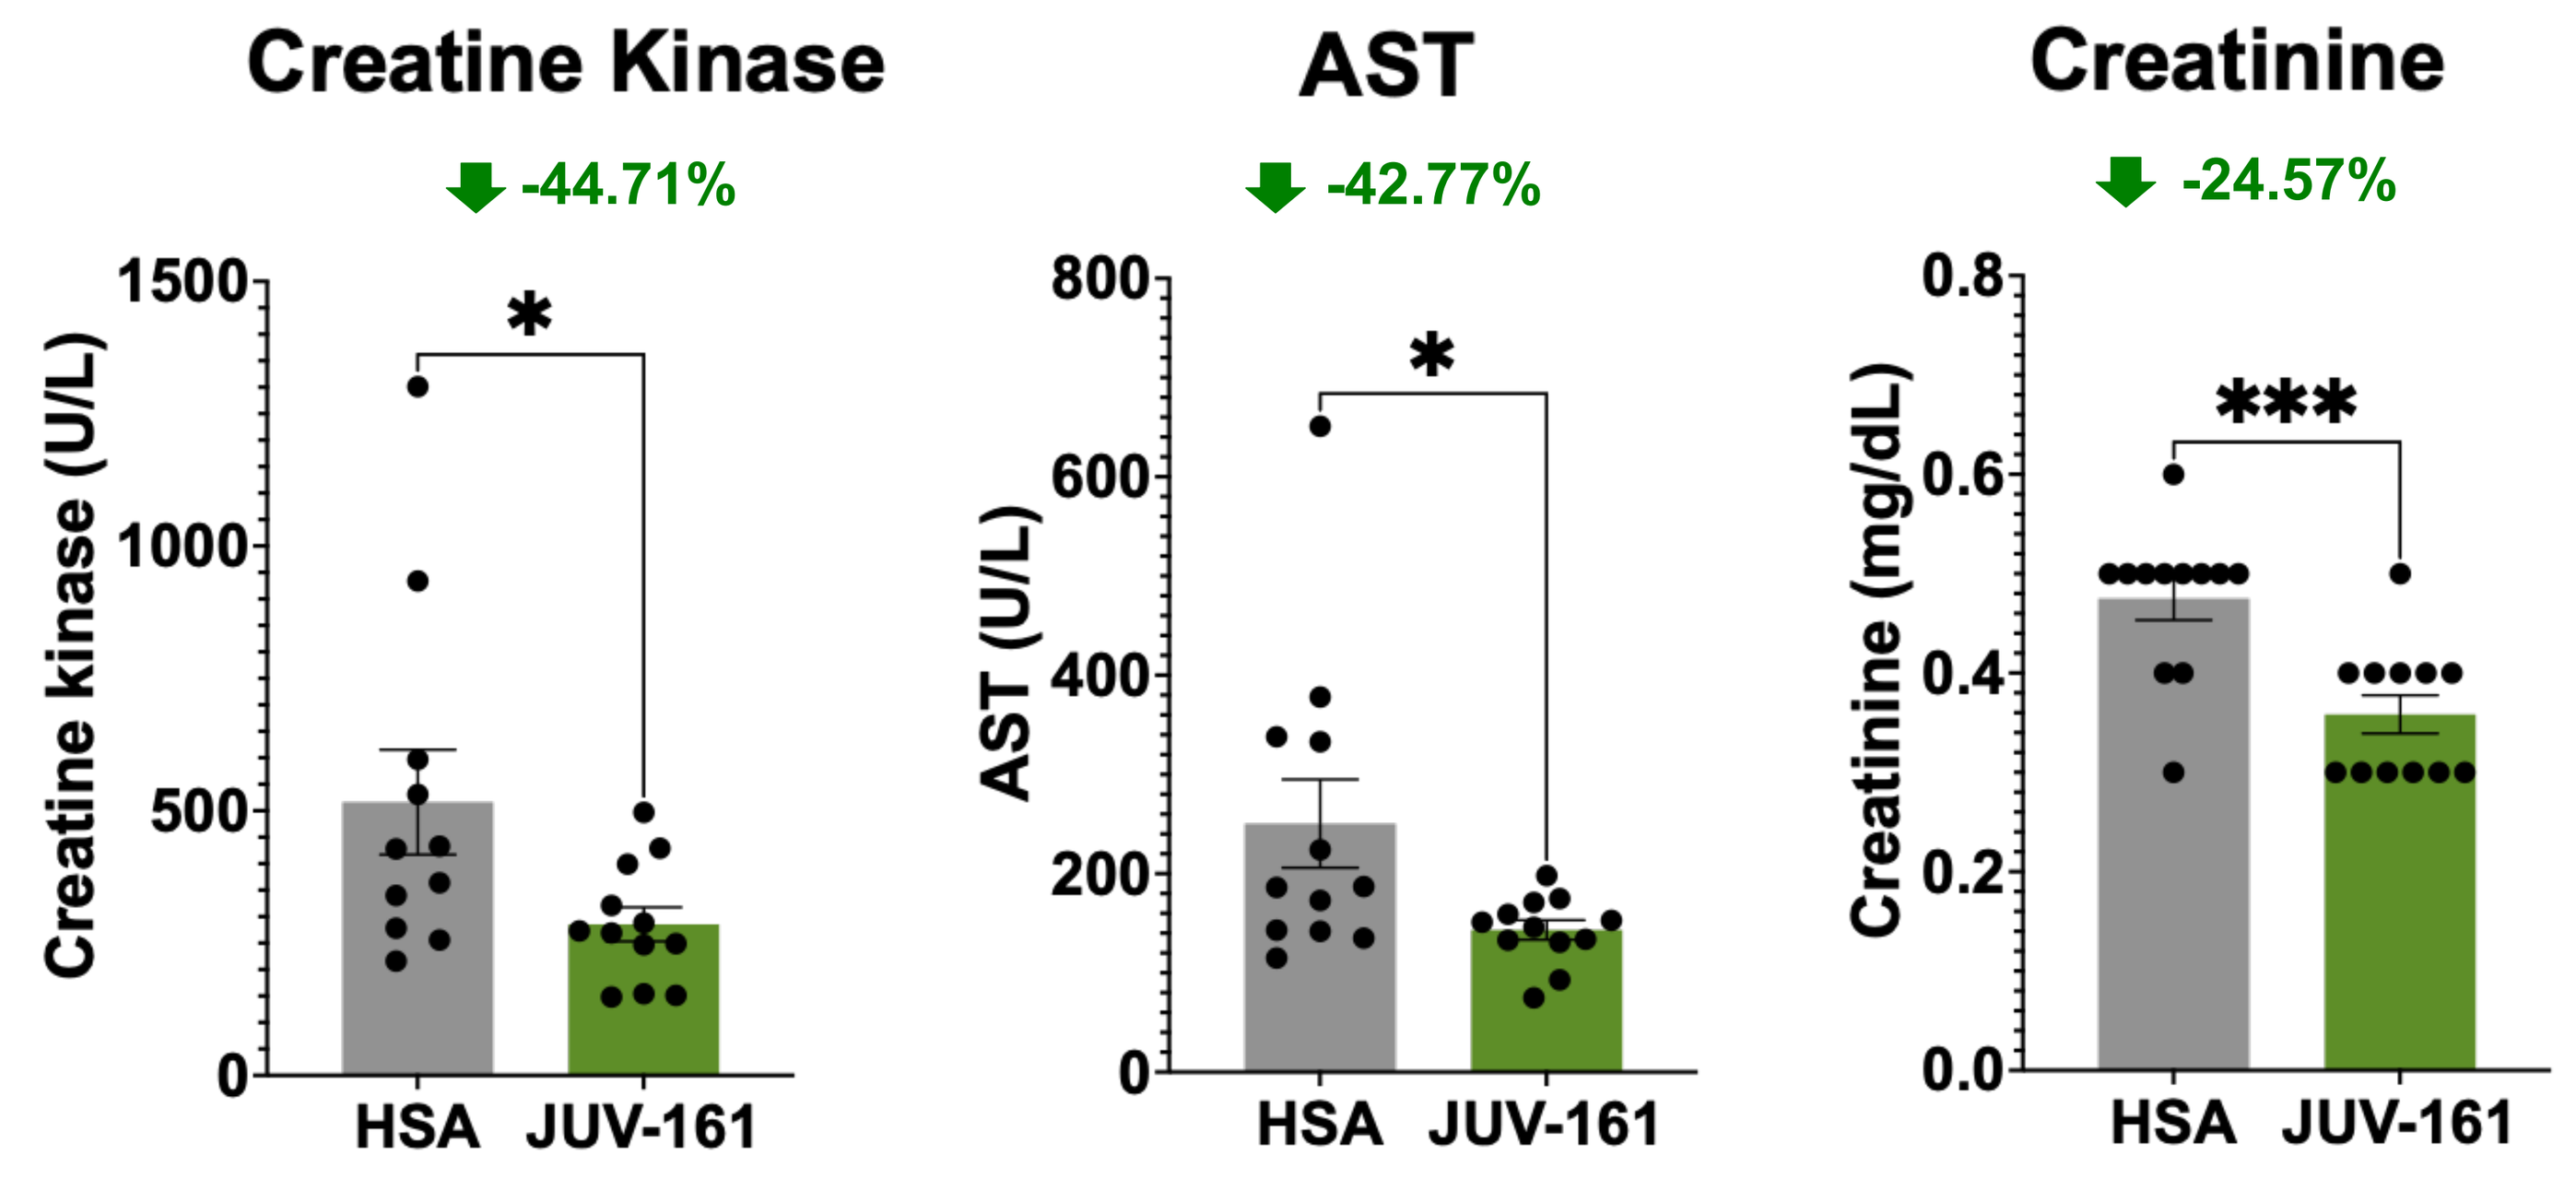
Creatine Kinase
-44.71%
Creatine kinase (U/L)
AST
-42.77%
AST (U/L)
Creatinine
-24.57%
Creatinine (mg/dL)
HSA
JUV-161

Figure 3: Chronic JUV-161 treatment improves systemic metabolic parameters in obese type 2 diabetic db/db mice. *At the end of the study, serum was collected and multiple parameters were assessed. Unpaired t-tests with Welch's correction. Statistical significance is indicated as follows: ***p < 0.001 & *p < 0.05. Sample size: n = 12 per group.*

JUV-161 improves lean/muscle mass in obese type 2 diabetic mice

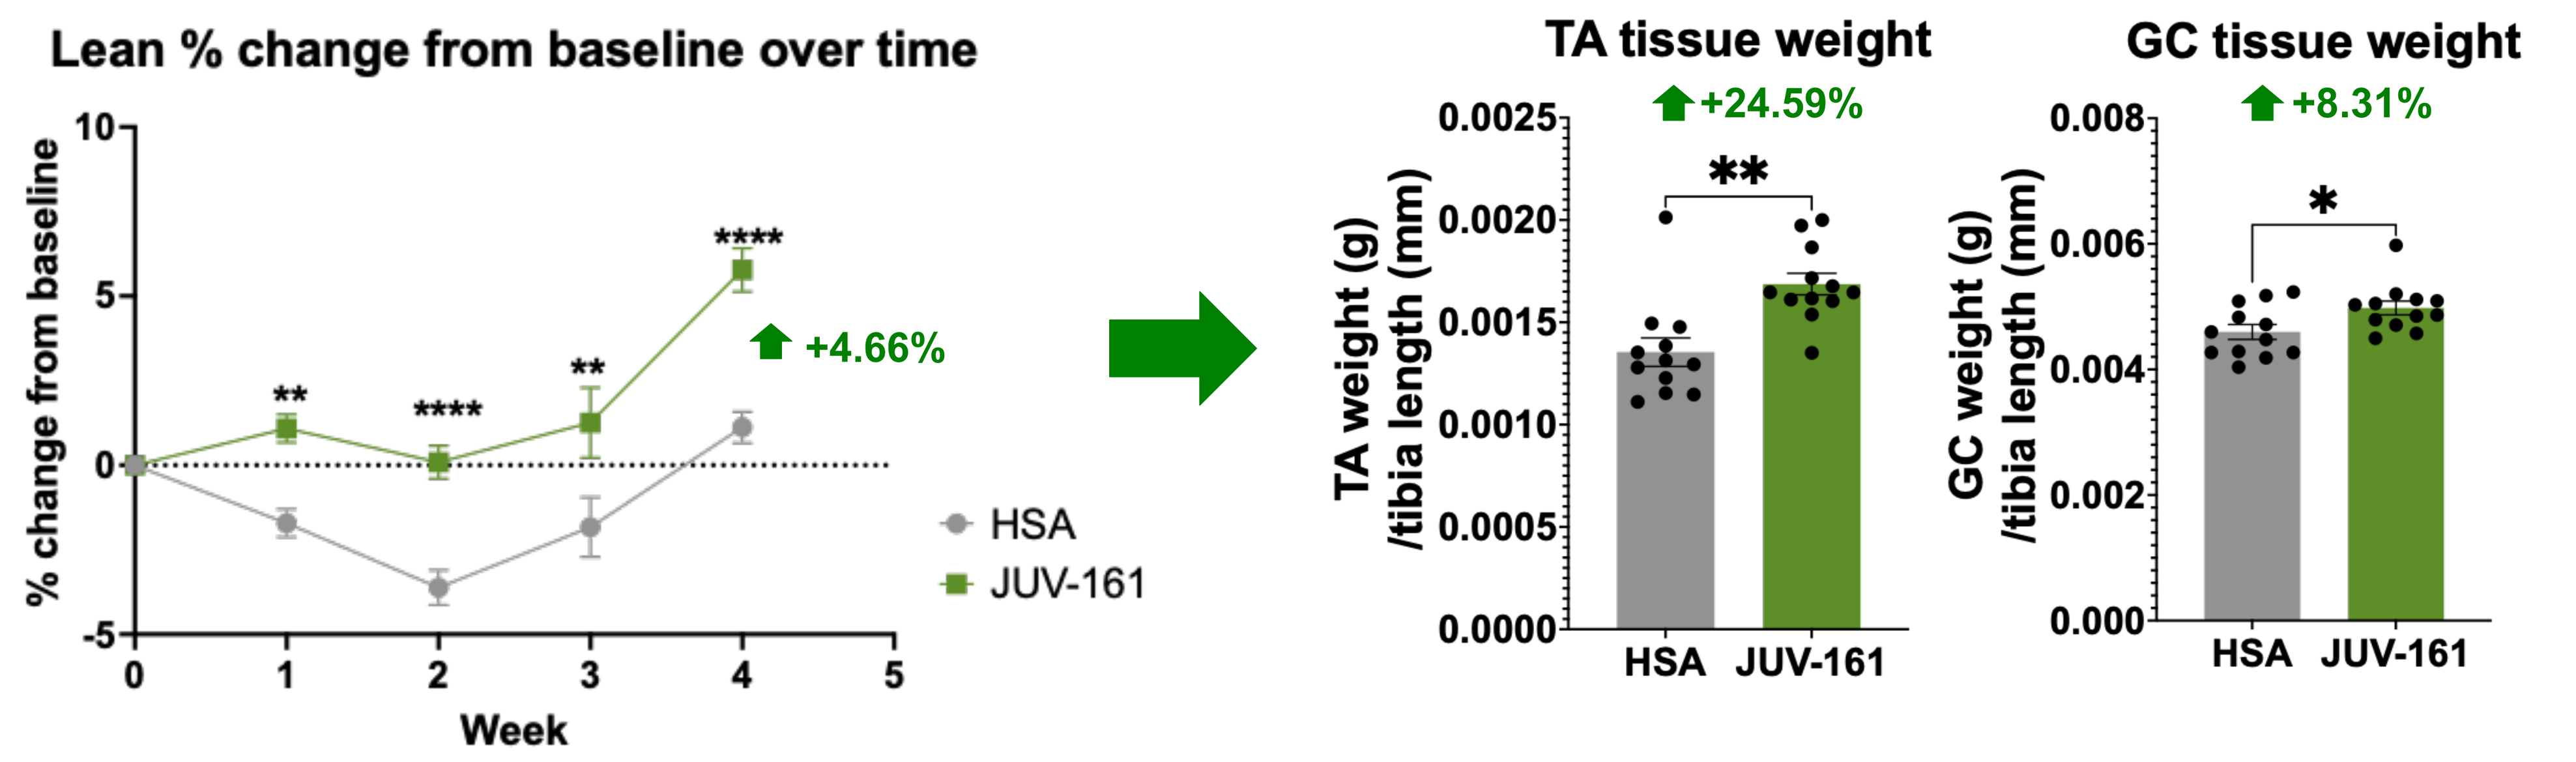
Lean % change from baseline over time
% change from baseline
Week
HSA
JUV-161
+4.66%
TA tissue weight
+24.59%
TA weight (g) /tibia length (mm)
GC tissue weight
+8.31%
GC weight (g) /tibia length (mm)
HSA
JUV-161

Figure 4: Chronic JUV-161 treatment increases lean mass (EchoMRI) and muscle tissue weight in obese type 2 diabetic db/db mice*. Lean mass was measured weekly throughout the study using EchoMRI. At the end of the study, following chronic JUV-161 treatment, skeletal muscles—including tibialis anterior (TA) and gastrocnemius (GC)—were harvested and weighed. Data are presented as mean ± SEM and analyzed using unpaired t-tests with Welch's correction. Statistical significance is indicated as follows: ****p < 0.0001; **p < 0.01; *p < 0.05.*

JUV-161 enhances muscle cross-sectional area in obese type 2 diabetic mice

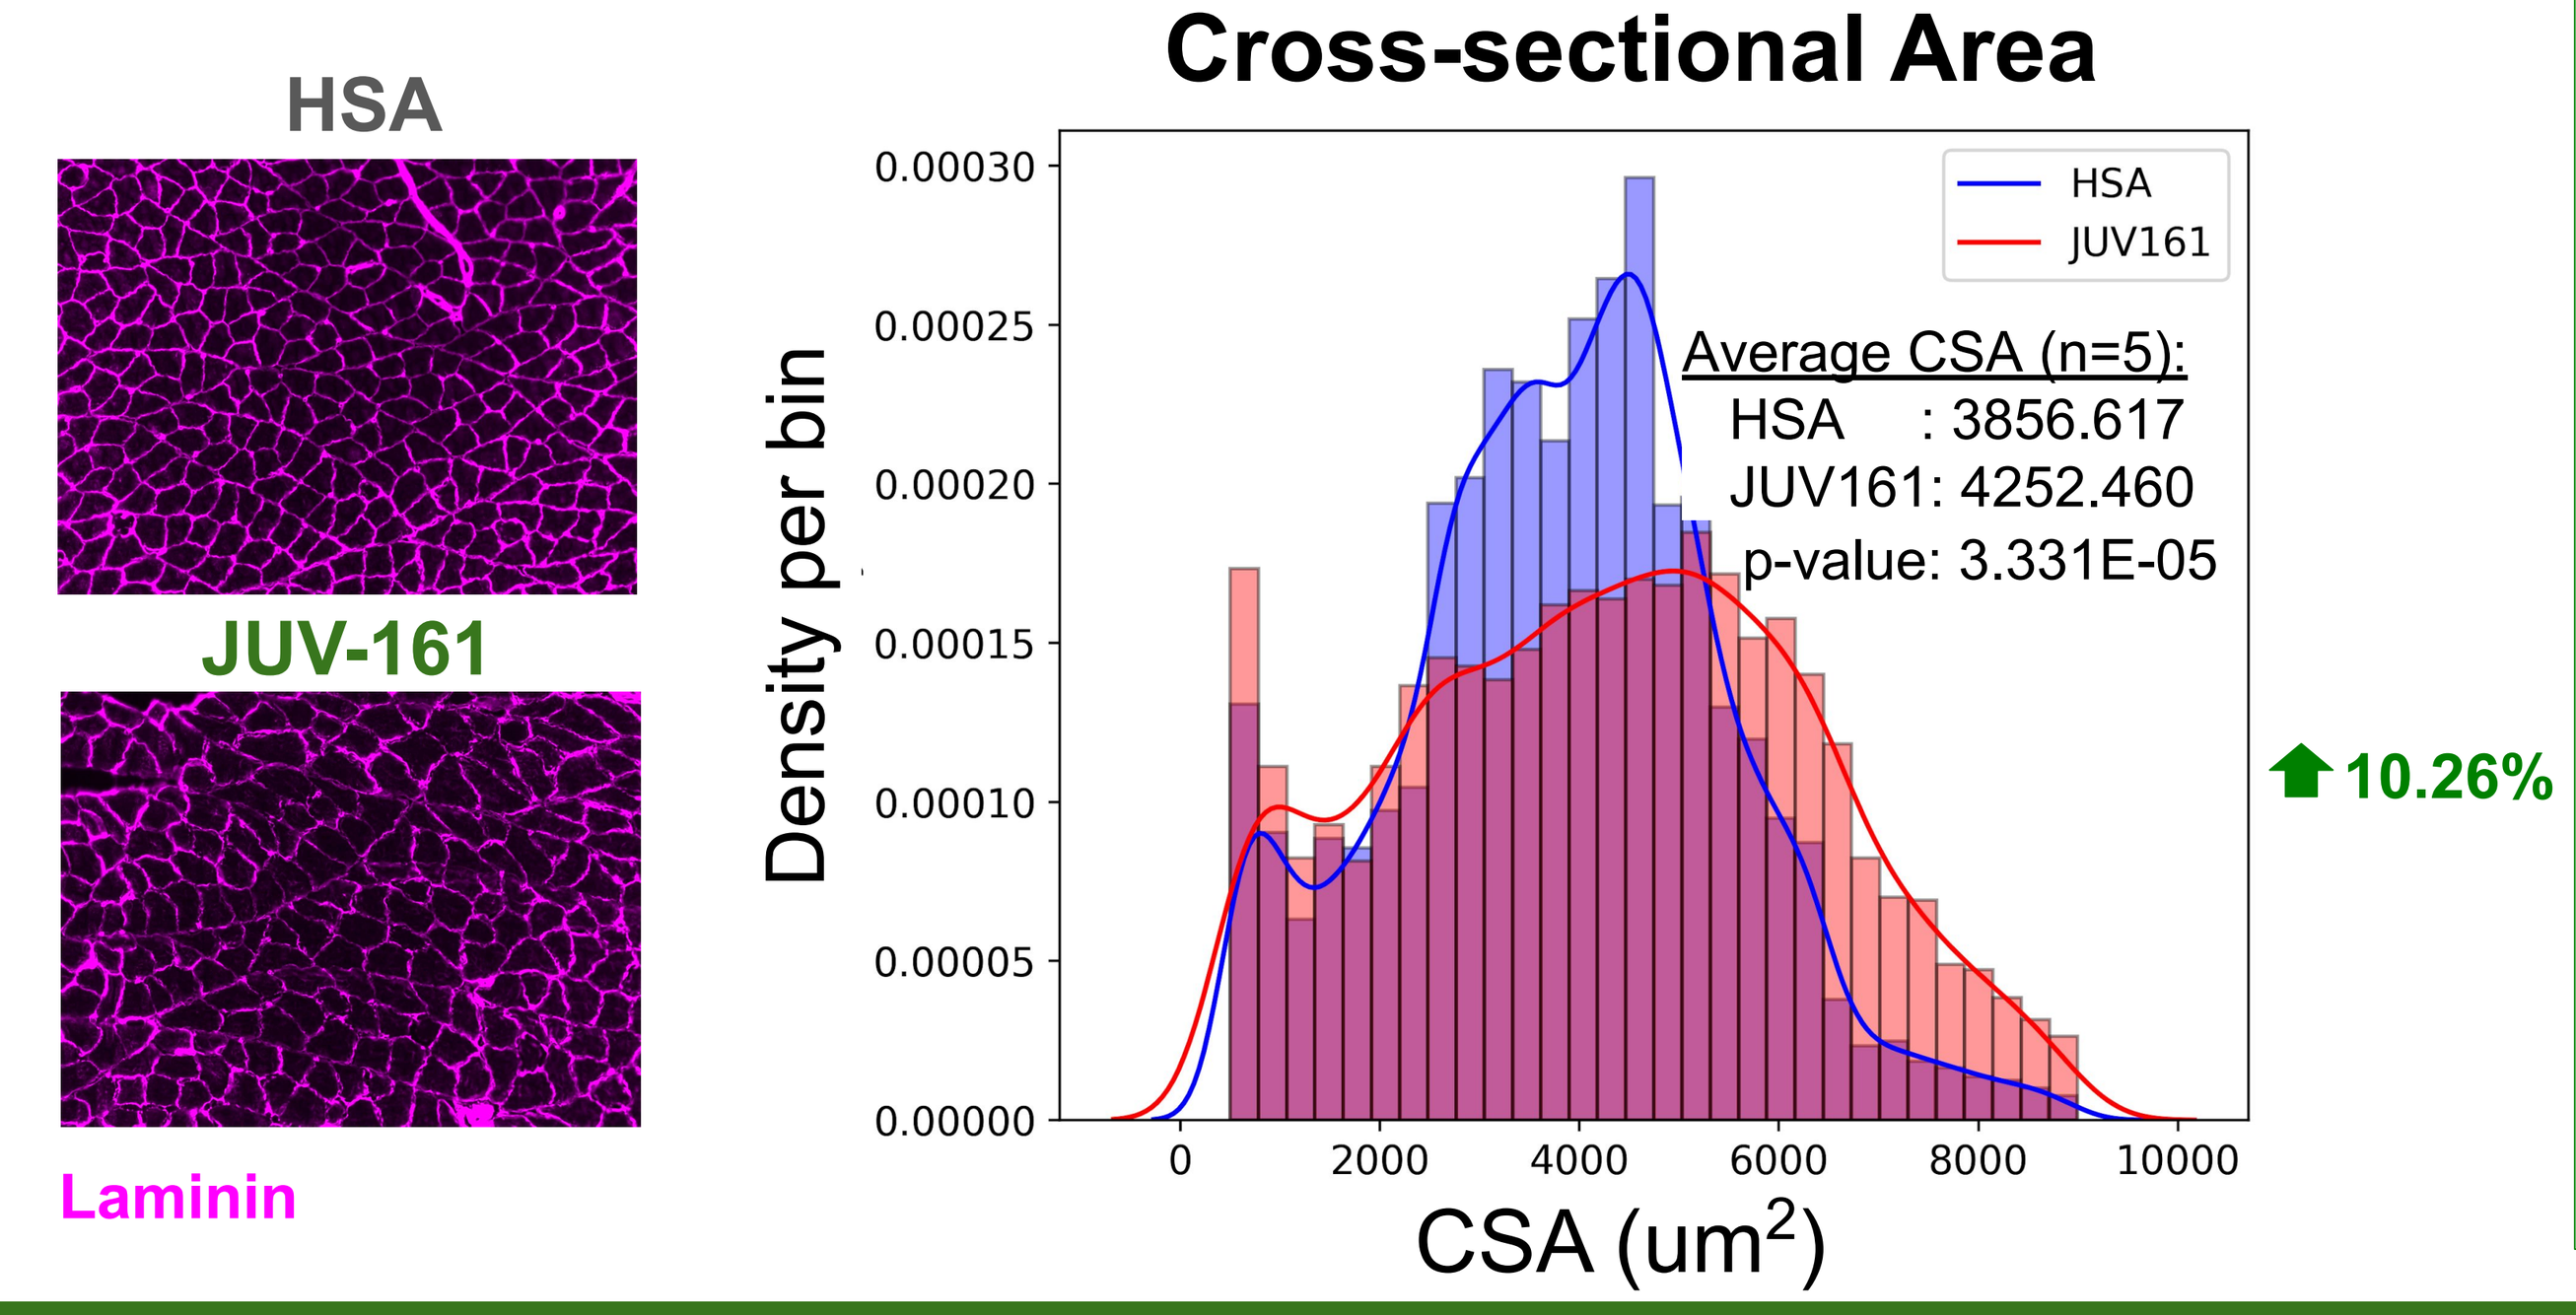
HSA
JUV-161
Laminin
Cross-sectional Area
Density per bin
HSA
JUV161
Average CSA (n=5):
HSA : 3856.617
JUV161: 4252.460
p-value: 3.331E-05
10.26%
CSA (um2)

Figure 5: Chronic JUV-161 treatment increases gastrocnemius (GC) muscle cross-sectional area in obese type 2 diabetic db/db mice. *At the end of the study, following chronic JUV-161 treatment, GC muscles were collected, sectioned, and analyzed for muscle fiber cross-sectional area. Data were analyzed using unpaired t-tests with Welch's correction. Statistical significance is indicated as follows: ****p < 0.0001*

Conclusions

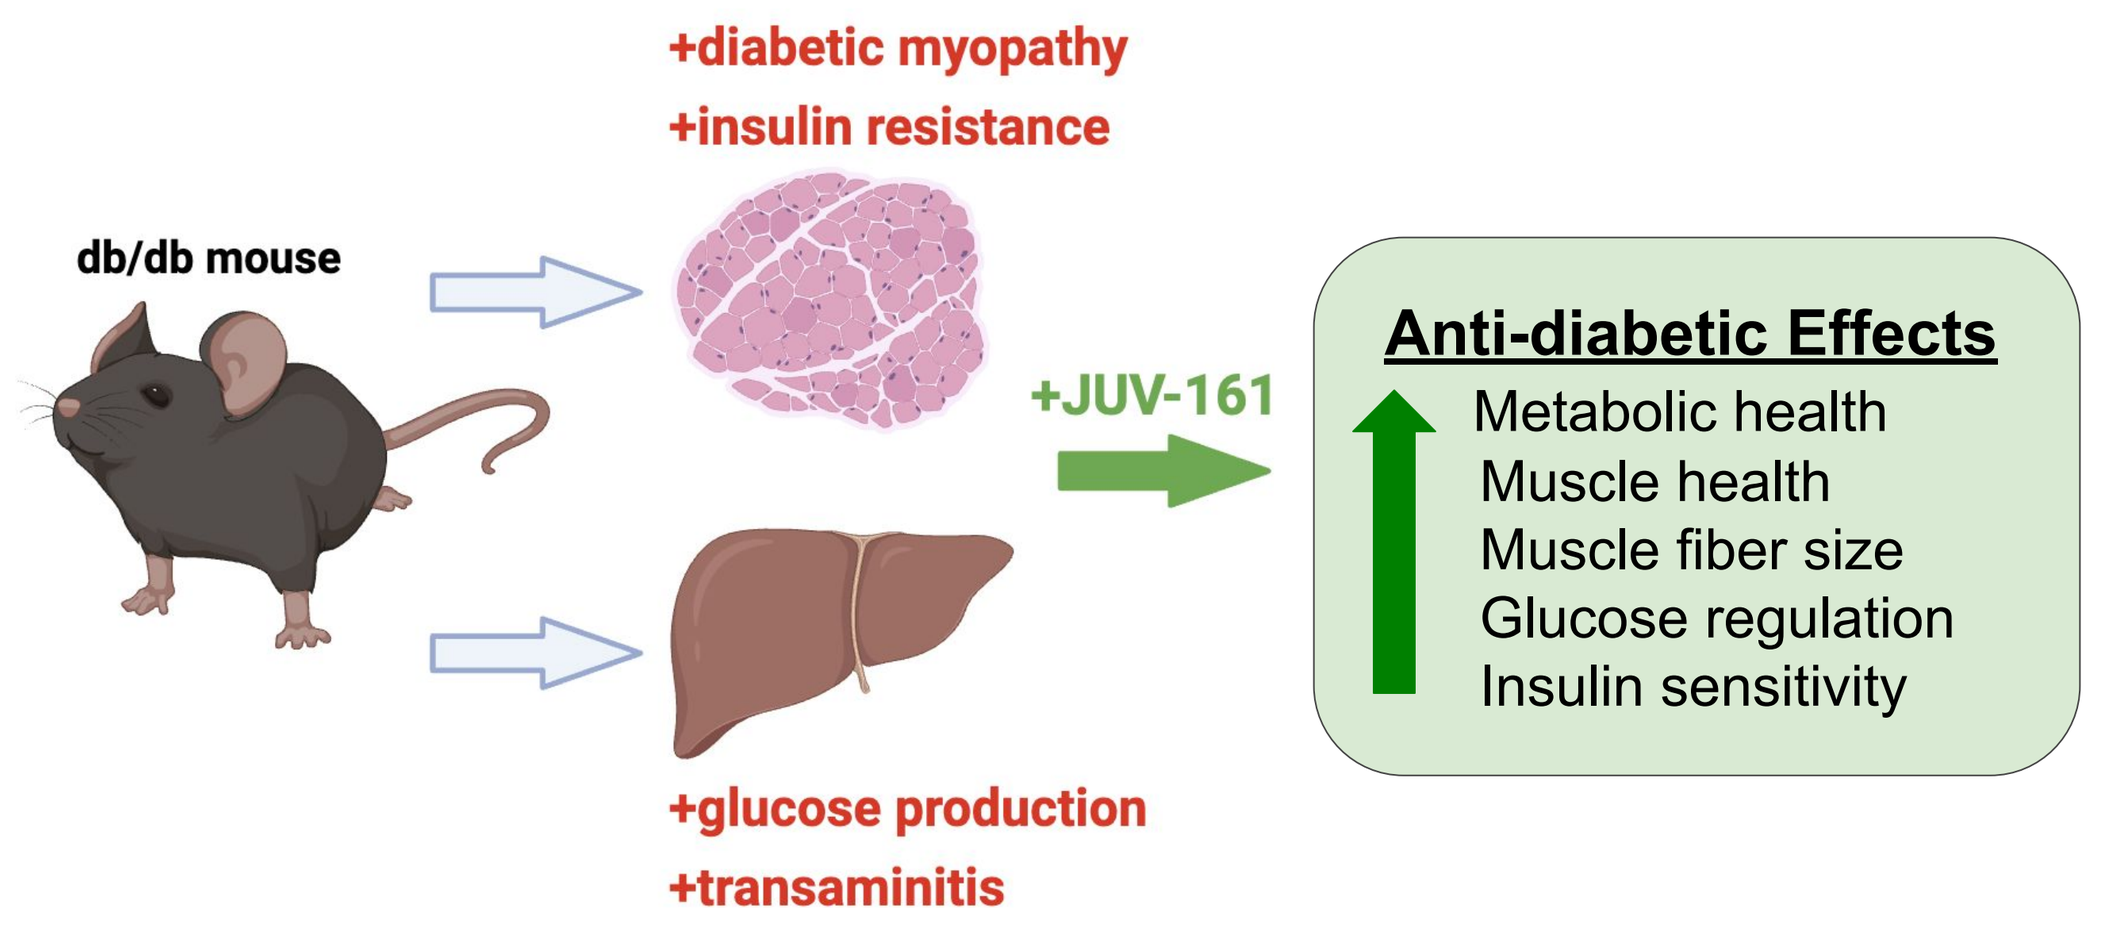
db/db mouse
+diabetic myopathy
+insulin resistance
+glucose production
+transaminitis
+JUV-161
Anti-diabetic Effects
Metabolic health
Muscle health
Muscle fiber size
Glucose regulation
Insulin sensitivity

- **JUV161 improved insulin sensitivity and modestly reduced HbA1c levels, indicating better glucose control.**
- **Increased lean muscle mass and TA/GC muscle size, supporting muscle health benefits.**
- **Significantly lowered serum AST, glucose, creatinine, and creatine kinase, reflecting improved metabolic and organ function.**
- **Overall, JUV161 shows promise as a type 2 diabetes therapy with added muscle-preserving and systemic benefits.**

Juvena Therapeutics, Inc., Redwood City, CA 94063. juvenatherapeutics.com

https://www.juvenatherapeutics.com